REX-8756 is an Oral, First-in-Class Orthosteric STAT6 Inhibitor That Demonstrates Efficacious Potential and Safety Differentiation in a Th2-Mediated Dermatitis Model

Patrick Metz, Howie Bregman, Rishi Vaswani, Kaylin Holderman, Jeong-Ho Kim, Jessica Ma, Angelo Moreno, Donglim Park, Daniel Treiber, Brian Hodous, Paul Smith, Ajay Nirula, Alexandra Gardino
Recludix Pharma, 3010 Science Park Road, San Diego, USA

BACKGROUND

Interleukins (IL)-4 and IL-13 are key drivers of the pathobiology underlying atopic dermatitis (AD), a chronic inflammatory skin disease. Systemic treatments for AD include biologics directly targeting the IL-4/IL-13 signaling pathway and oral Janus Kinase (JAK) inhibitors. Biologics require injections, and JAK inhibition can lead to broad immunosuppression and hematologic side effects. Selective oral STAT6 inhibition therefore represents an opportunity to phenocopy the biologic efficacy of anti-IL-4Rα/IL-13 monoclonal antibodies, while avoiding known JAK-mediated adverse events.

The SH2 domain is the exclusive mediator of STAT6 binding to the IL-4/13 cytokine receptors and is required to trigger downstream transcriptional activity. Orthosteric inhibition therefore represents an optimal strategy for modulating activity of this attractive therapeutic target. Recludix has an innovative SH2 domain platform generating small molecule inhibitors of these previously undruggable protein domains.

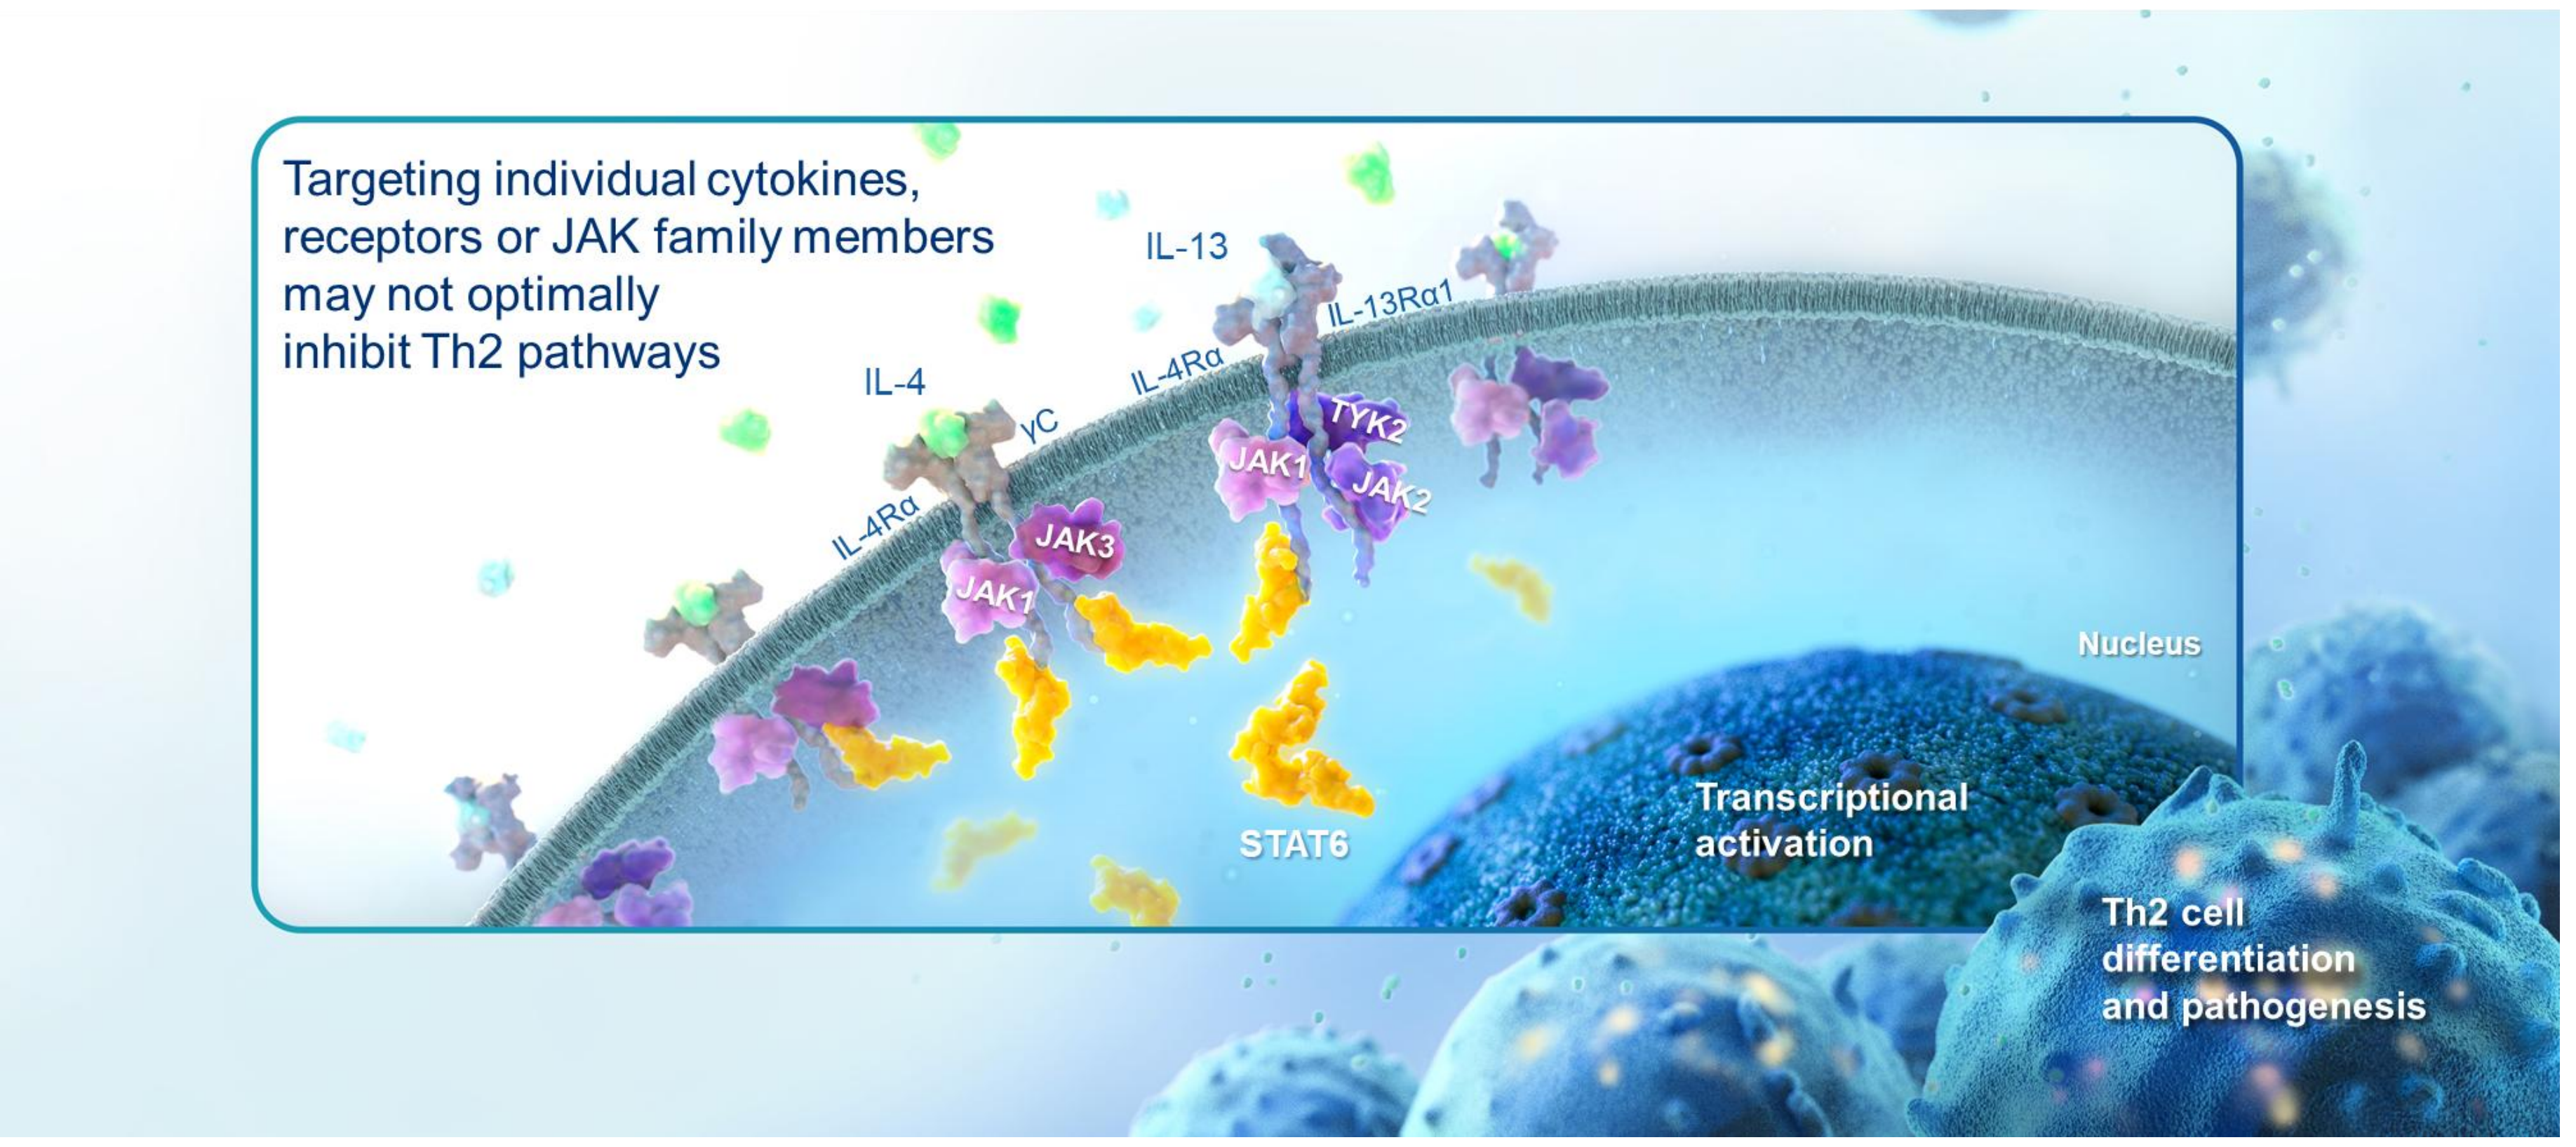
Targeting individual cytokines, receptors or JAK family members may not optimally inhibit Th2 pathways
IL-13
IL-13Rα1
IL-4
IL-4Rα
γC
IL-4Rα
TYK2
JAK1
JAK2
JAK3
JAK1
Nucleus
STAT6
Transcriptional activation
Th2 cell differentiation and pathogenesis

Figure 1. Inhibiting STAT6 to selectively target Th2 inflammatory disease pathways

METHODS & TECHNOLOGY

The Recludix Pharma SH2 domain discovery platform integrates custom and proprietary DNA-encoded libraries (DEL), structure-based design enabled by rapid STAT SH2 domain crystallography, and a biochemical assay panel to assess selectivity and potency (Figure 2).

.

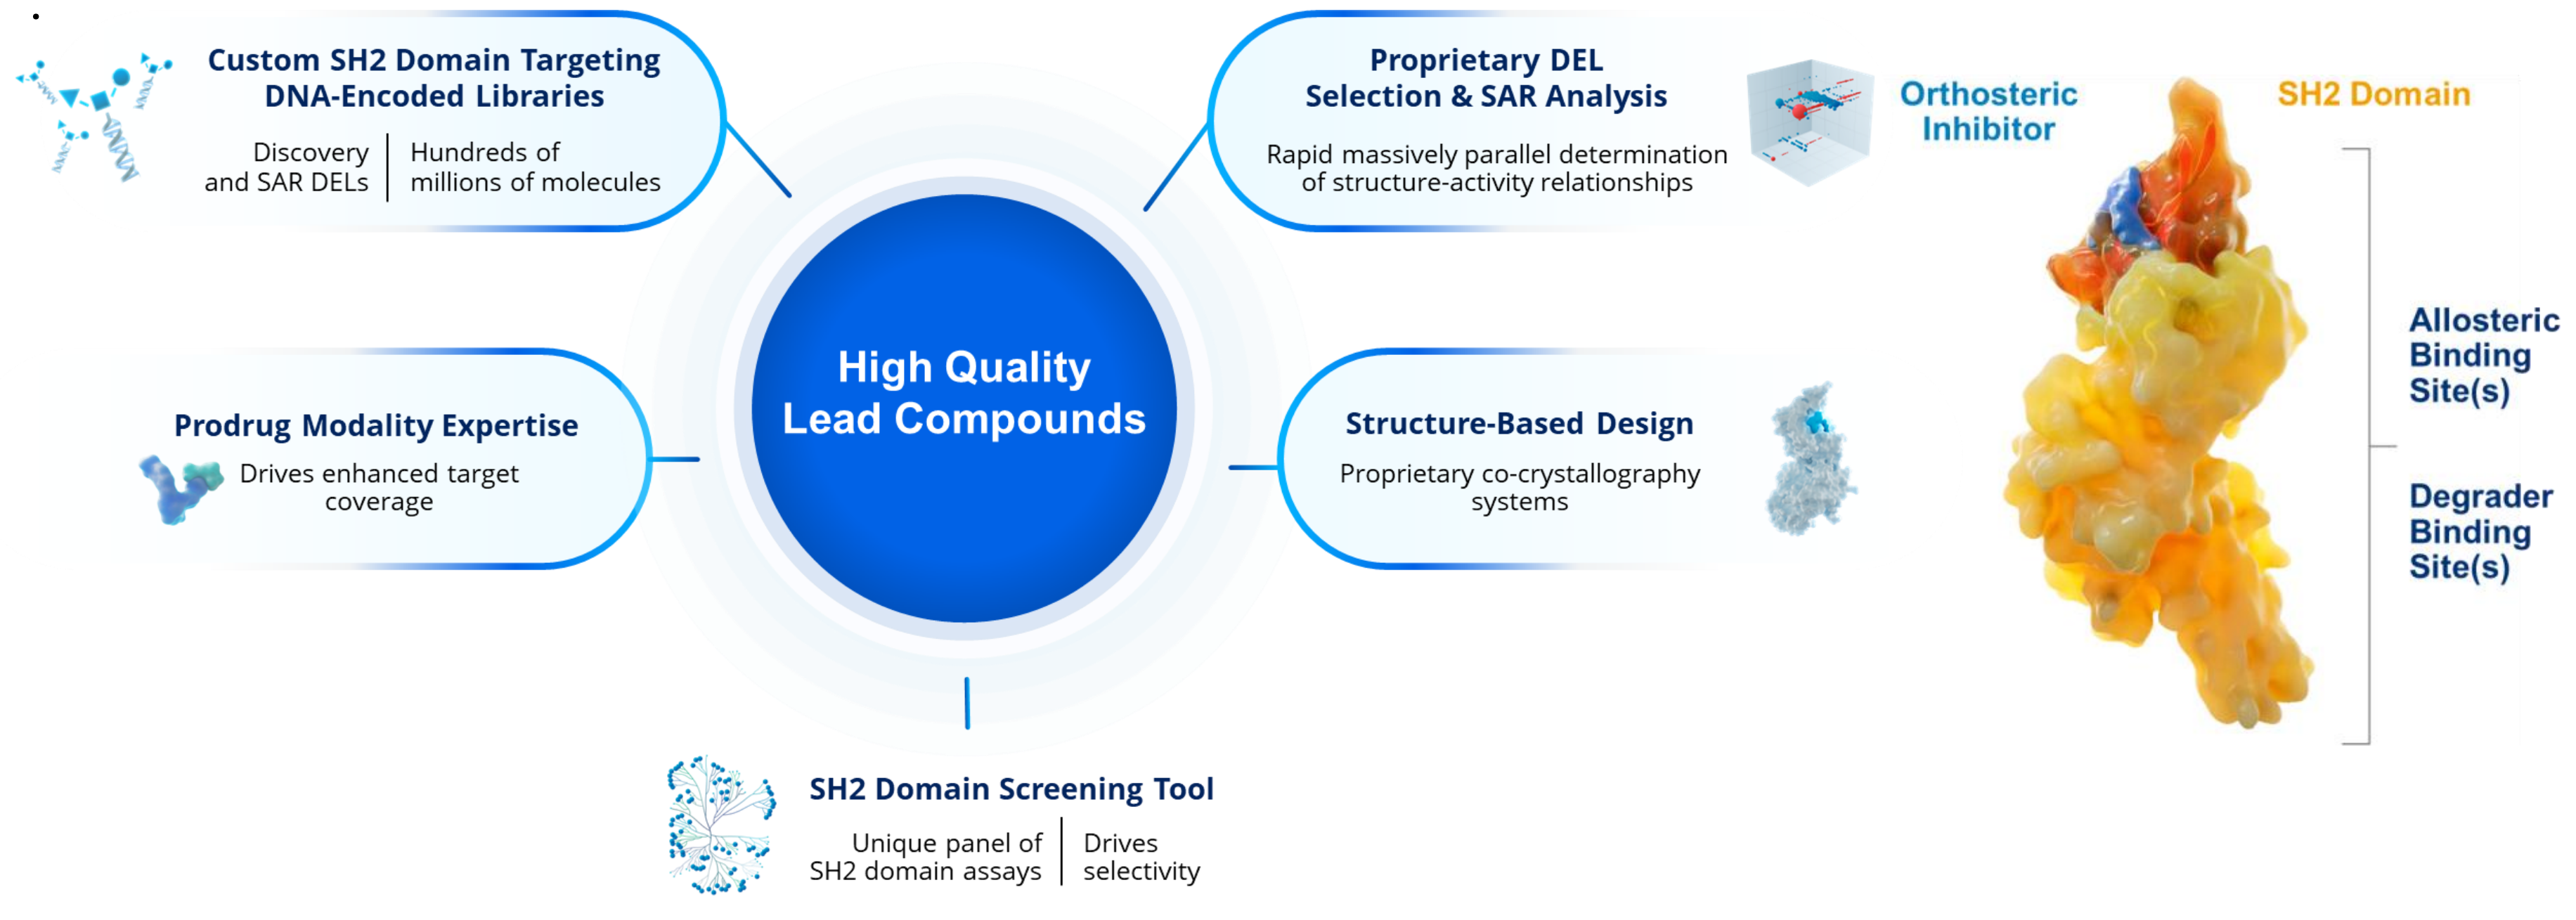
Custom SH2 Domain Targeting DNA-Encoded Libraries
Discovery and SAR DELs
Hundreds of millions of molecules
Proprietary DEL Selection & SAR Analysis
Rapid massively parallel determination of structure-activity relationships
Orthosteric Inhibitor
SH2 Domain
High Quality Lead Compounds
Prodrug Modality Expertise
Drives enhanced target coverage
Structure-Based Design
Proprietary co-crystallography systems
Allosteric Binding Site(s)
Degrader Binding Site(s)
SH2 Domain Screening Tool
Unique panel of SH2 domain assays
Drives selectivity

Figure 2. Integrated SH2 domain drug discovery and chemical optimization platform

Proprietary DEL libraries identify novel hits which feed into crystallography for rapid hit optimization using structure-based drug design, in silico optimization, and a suite of biochemical and biophysical assays to monitor potency and physicochemical properties. Discovery of potent and selective STAT6 inhibitors exemplifies the value of this integrated approach to target previously undruggable targets.

DATA RESULTS

Recludix has identified the first known STAT6 orthosteric SH2 domain inhibitors exemplified by REX-8756 that demonstrates exquisite potency and selectivity across a panel covering roughly 75% of human SH2 domains (Table 1). Treatment of REX-8756 results in complete blockade of the IL-4 and IL-13 signaling pathway as demonstrated by the full inhibition of phospho-STAT6 and disruption of TARC production which is transcriptionally regulated by STAT6 and is a known biomarker for type 2 inflammatory diseases, following stimulation of human PBMCs with IL-4 or IL-13 (Figure 3).

	Biochemical potency (SH2scan K_D)	Cellular potency (pSTAT6 IC_{50} in human PBMCs)	Biochemical STAT family selectivity	Cellular selectivity (PBMCs)	SH2 domain selectivity
REX-8756	0.033 nM	0.72 nM (IL-4) 0.19 nM (IL-13)	>1,000X vs. STAT1/2/3/4/5	>1,000X vs. STAT1/2/3/4/5	

Table 1. Biochemical and cellular potency and selectivity of orthosteric SH2 domain inhibitor REX-8756

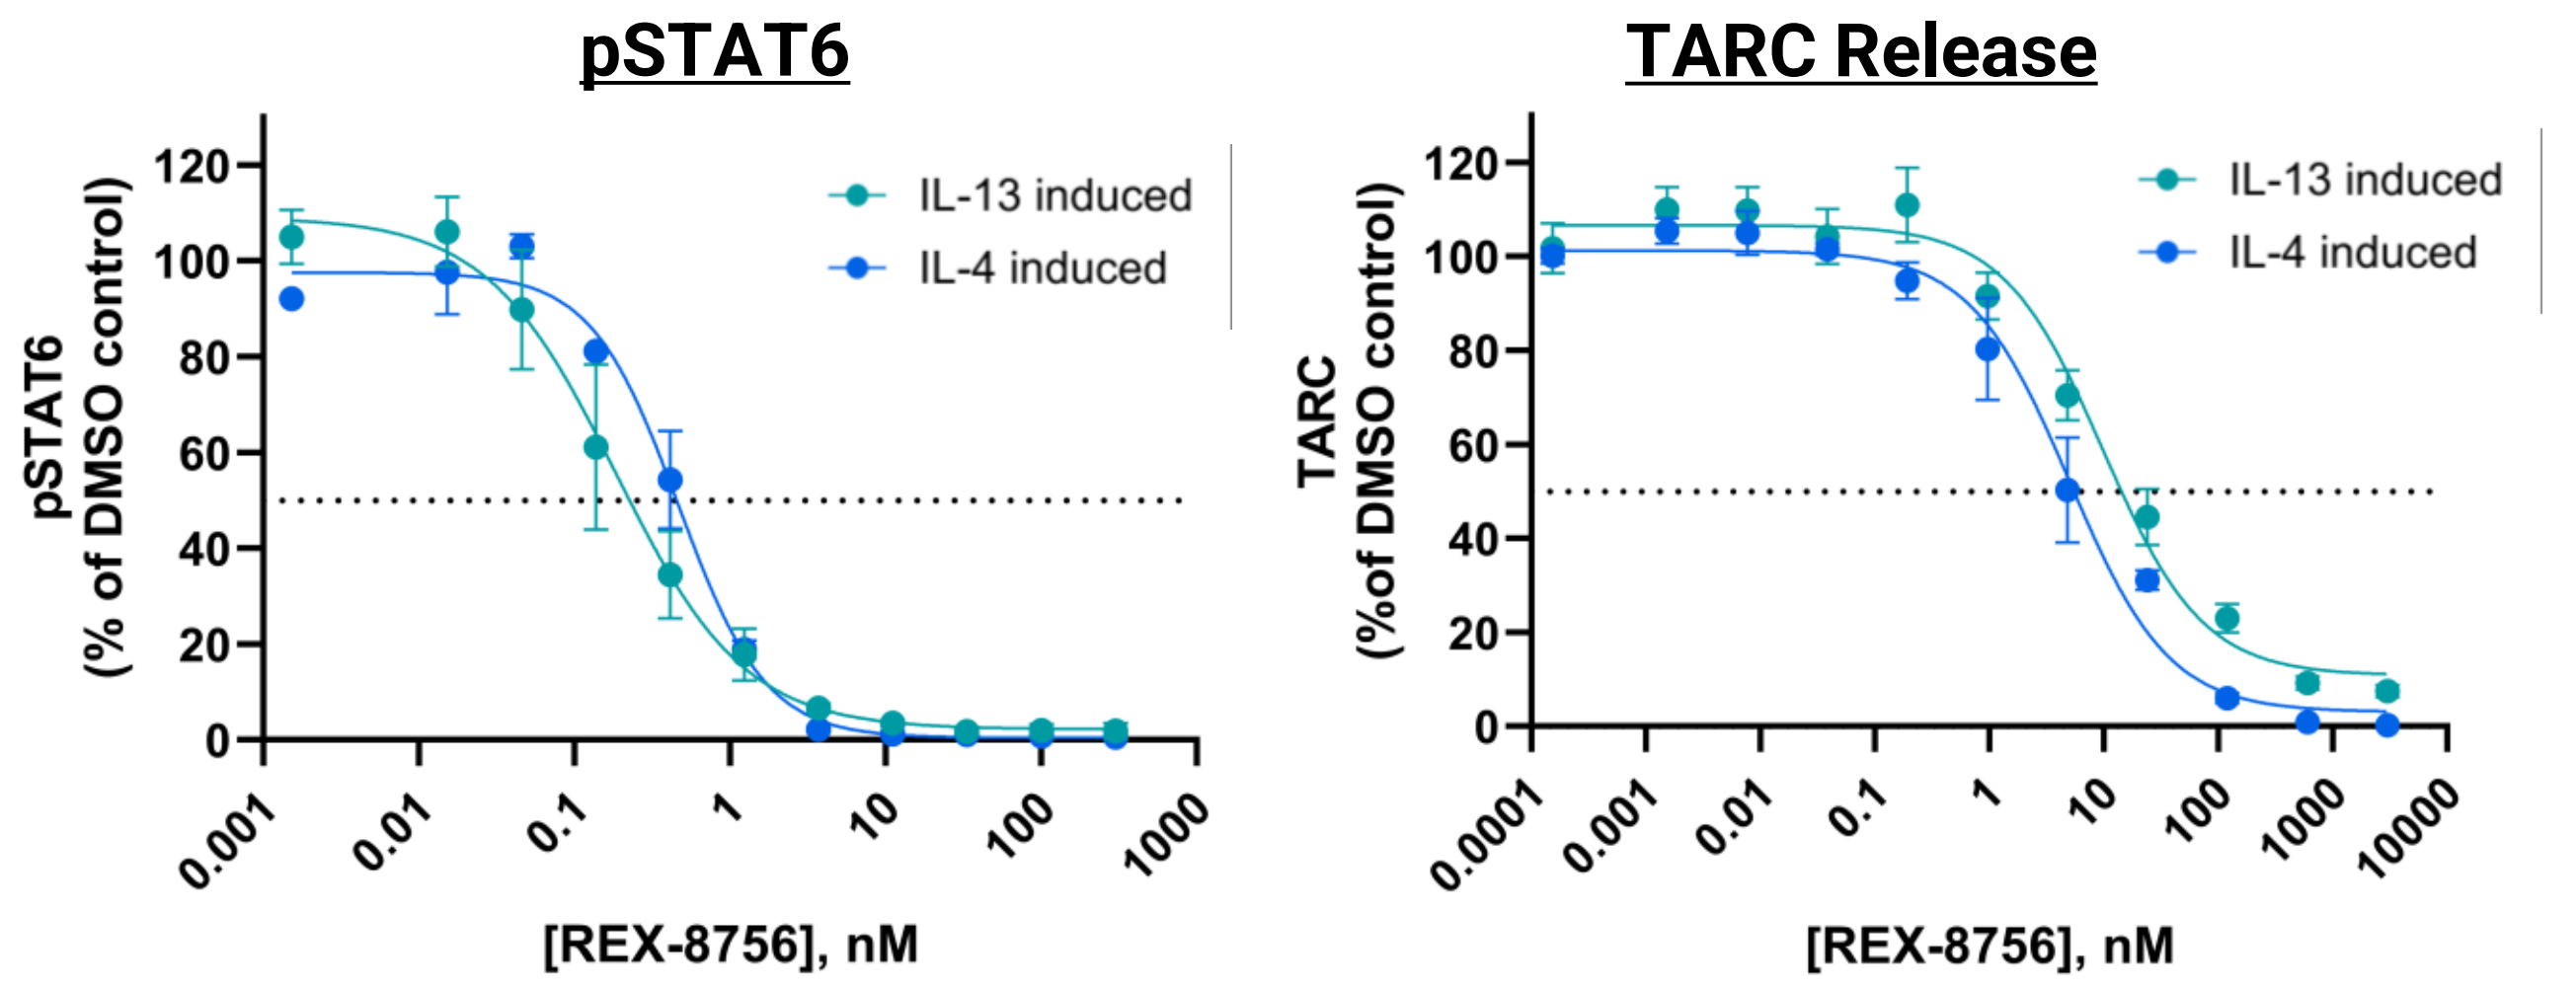
pSTAT6
pSTAT6 (% of DMSO control)
IL-13 induced
IL-4 induced
[REX-8756], nM
TARC Release
TARC (%of DMSO control)
IL-13 induced
IL-4 induced
[REX-8756], nM

Figure 3. Orthosteric SH2 domain inhibitor REX-8756 blocks IL-4 and IL-13 signaling pathway

The effect of REX-8756 on Th cell differentiation into various functional subtypes (Th0, Th1, Th2, and TH17) in human PBMCs and STAT5-driven activation of luciferase expression initiated by EPO or TPO was evaluated and compared to JAK family inhibitors and dupilumab to assess risks of broad immunosuppression and hematologic side effects.

REX-8756 phenocopies dupilumab and differentiates from JAK inhibitors by selectively blocking T helper cell differentiation into the type 2 (Th2) subtype without impacting hematologic homeostasis (Table 2).

		T CELL FUNCTION				HEMATOLOGIC HOMEOSTASIS	
		General adaptive immune response	Defense against viruses and bacteria	Defense against extracellular pathogens	Defense against parasites and mediation of antibody responses	Erythropoiesis	Thrombopoiesis
		T Cell Activation (CD25)	Th1 Cell Function (IFNγ)	Th17 Cell Function (IL-17A)	Th2 Cell Function (IL-5)	EPO-Induced STAT5-Driven Transcription	TPO-Induced STAT5-Driven Transcription
STAT6 Inhibitor	REX-8756	>1,000 nM	>1,000 nM	>1,00 nM	19 nM	>10,000 nM	>10,000 nM
IL-4 / IL-13 Antagonist	Dupilumab	>1,000 nM	>1,000 nM	>1,000 nM	22 nM	>1,000 nM	>1,000 nM
JAK Inhibitors	Abrocitinib	1,300 nM	900 nM	73 nM	81 nM	3,100 nM	2,800 nM
	Upadacitinib	39 nM	36 nM	7.4 nM	4.4 nM	69 nM	20 nM
	Baricitinib	110 nM	210 nM	25 nM	15 nM	55 nM	43 nM

Selectivity relative to Th2 inhibition: >30X 10-30X <10X

Table 2. REX-8756 demonstrates biologic-like selective Th2 inhibition and differentiates from JAK inhibitors in functional T helper differentiation and hematologic homeostasis assyas. Average IC_{50} values are summarized for each assay.

In a preclinical model of atopic dermatitis, blocking STAT6 activation with REX-8756 attenuated skin inflammation with efficacy comparable to antibody-mediated blockade of IL-4/13 pathways. Preclinical efficacy was also associated with reductions of Th2 cytokines (IL-13, IL-5) within the immune compartment (Figure 4).

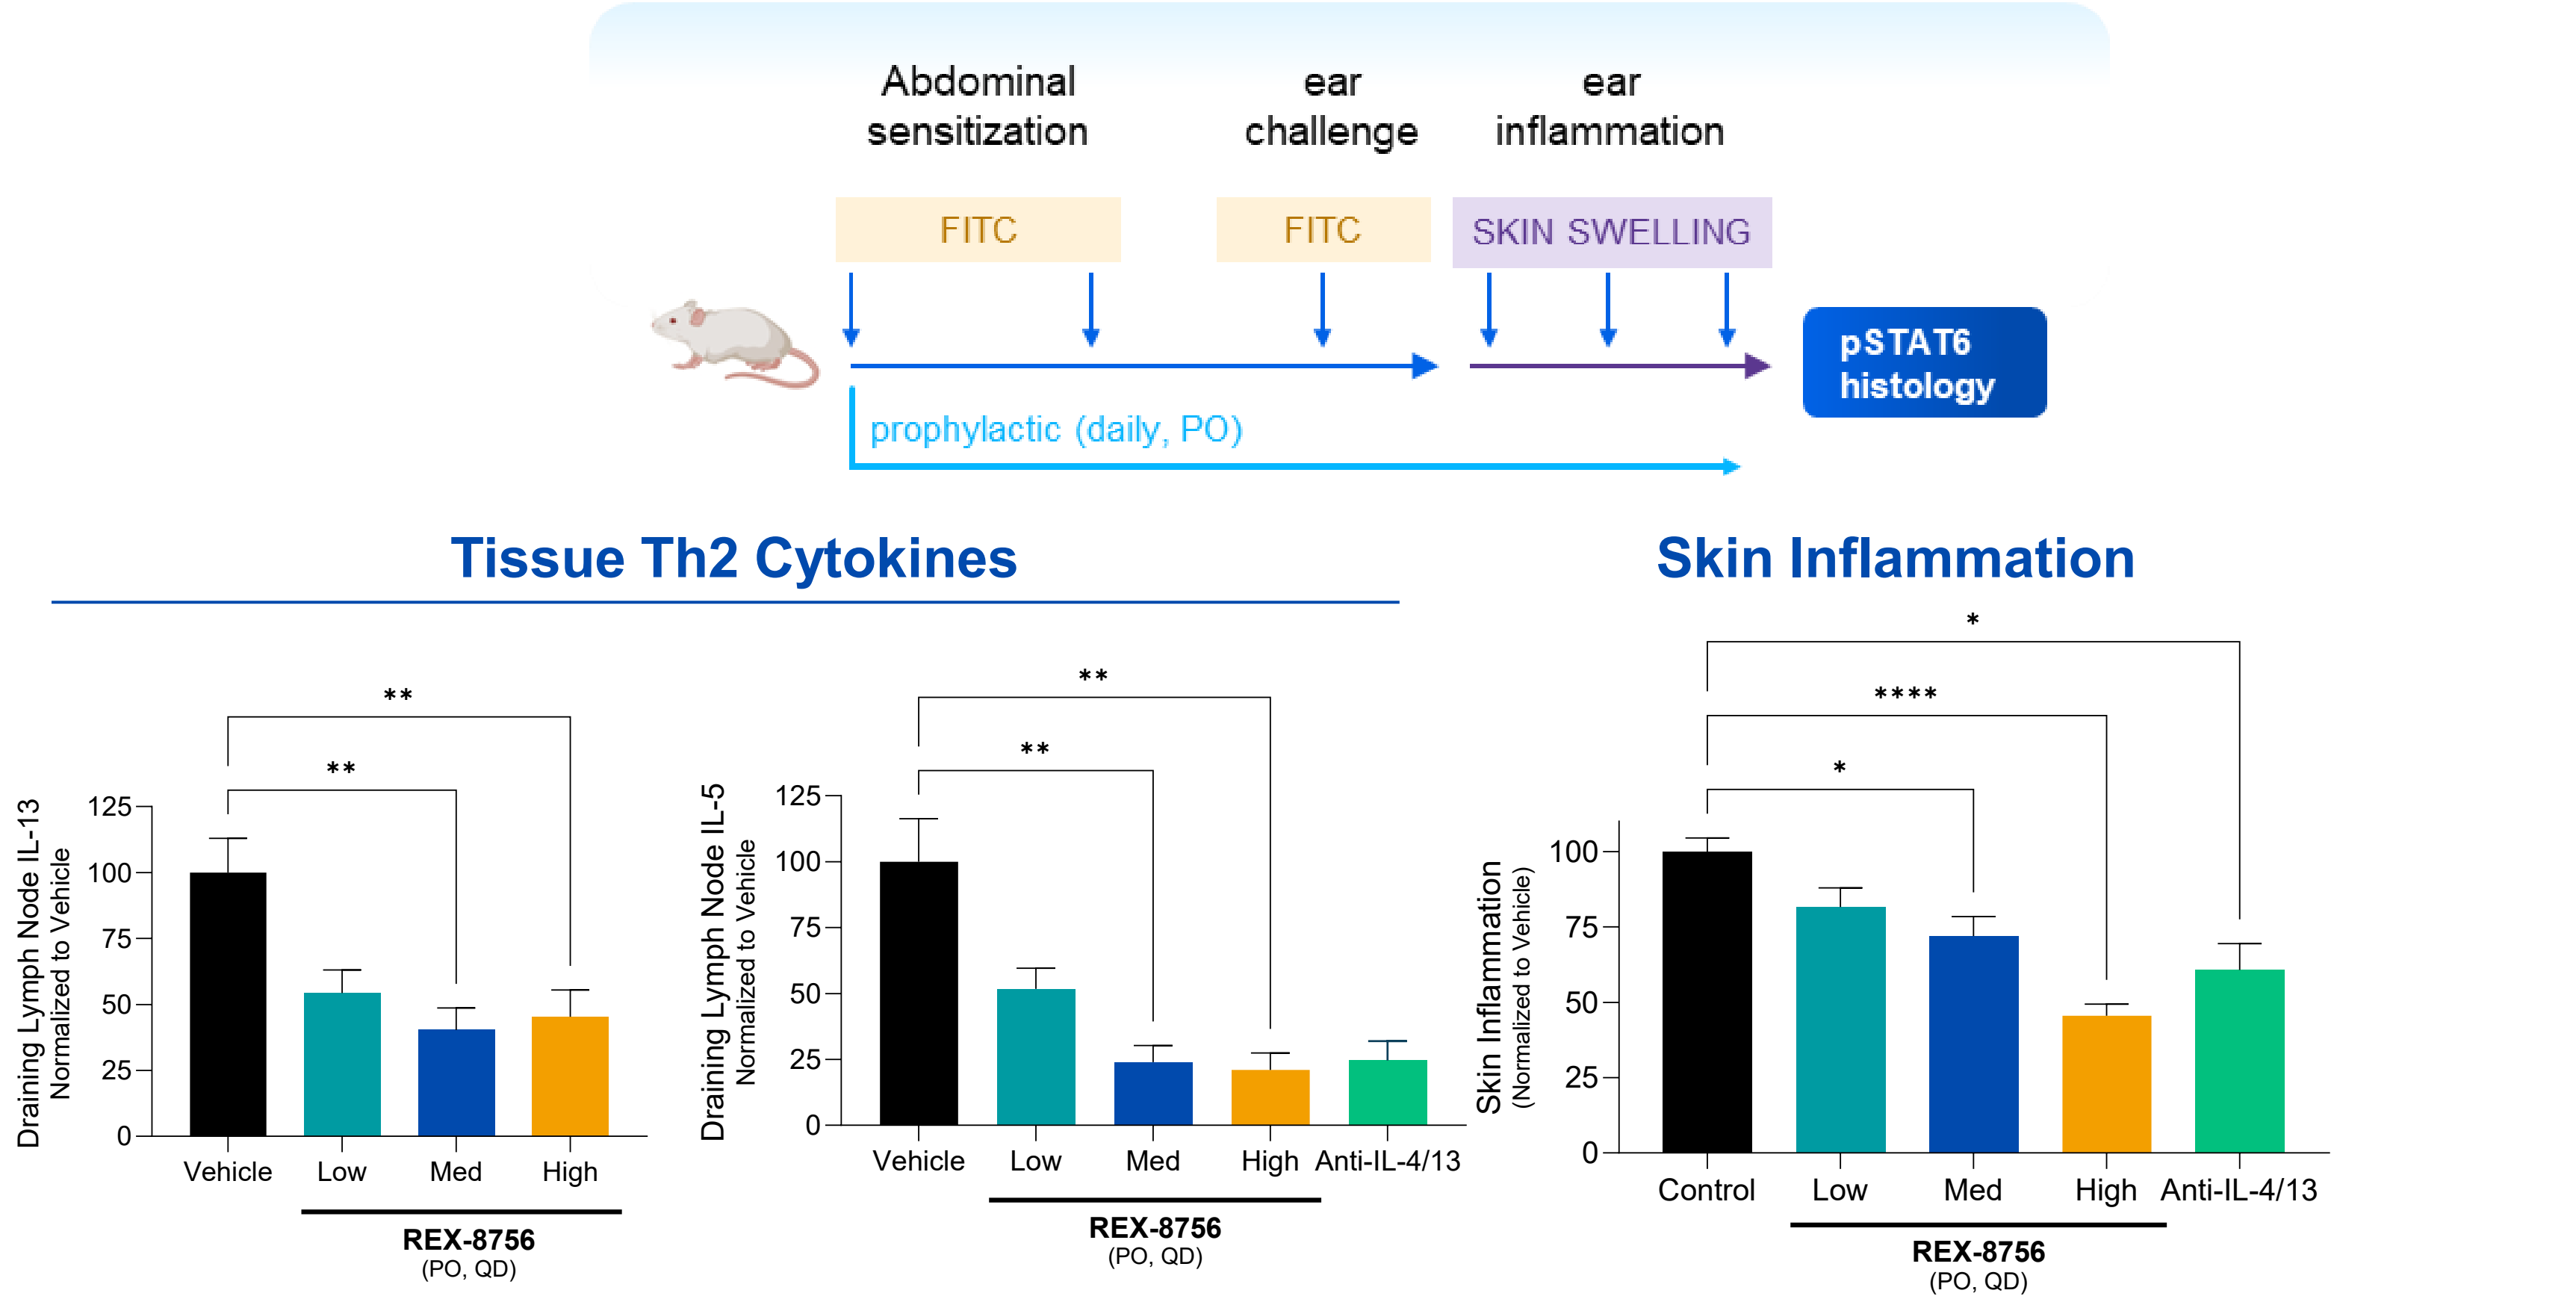
Abdominal sensitization
ear challenge
ear inflammation
FITC
FITC
SKIN SWELLING
pSTAT6 histology
prophylactic (daily, PO)
Tissue Th2 Cytokines
Skin Inflammation
Draining Lymph Node IL-13 Normalized to Vehicle
Vehicle
Low
Med
High
REX-8756 (PO, QD)
Draining Lymph Node IL-5 Normalized to Vehicle
Vehicle
Low
Med
High
Anti-IL-4/13
REX-8756 (PO, QD)
Skin Inflammation (Normalized to Vehicle)
Control
Low
Med
High
Anti-IL-4/13
REX-8756 (PO, QD)

Figure 4. REX-8756 inhibits tissue Th2 immunology and achieved comparable efficacy to combined anti-IL4/IL-13 biologics in a dermatitis model

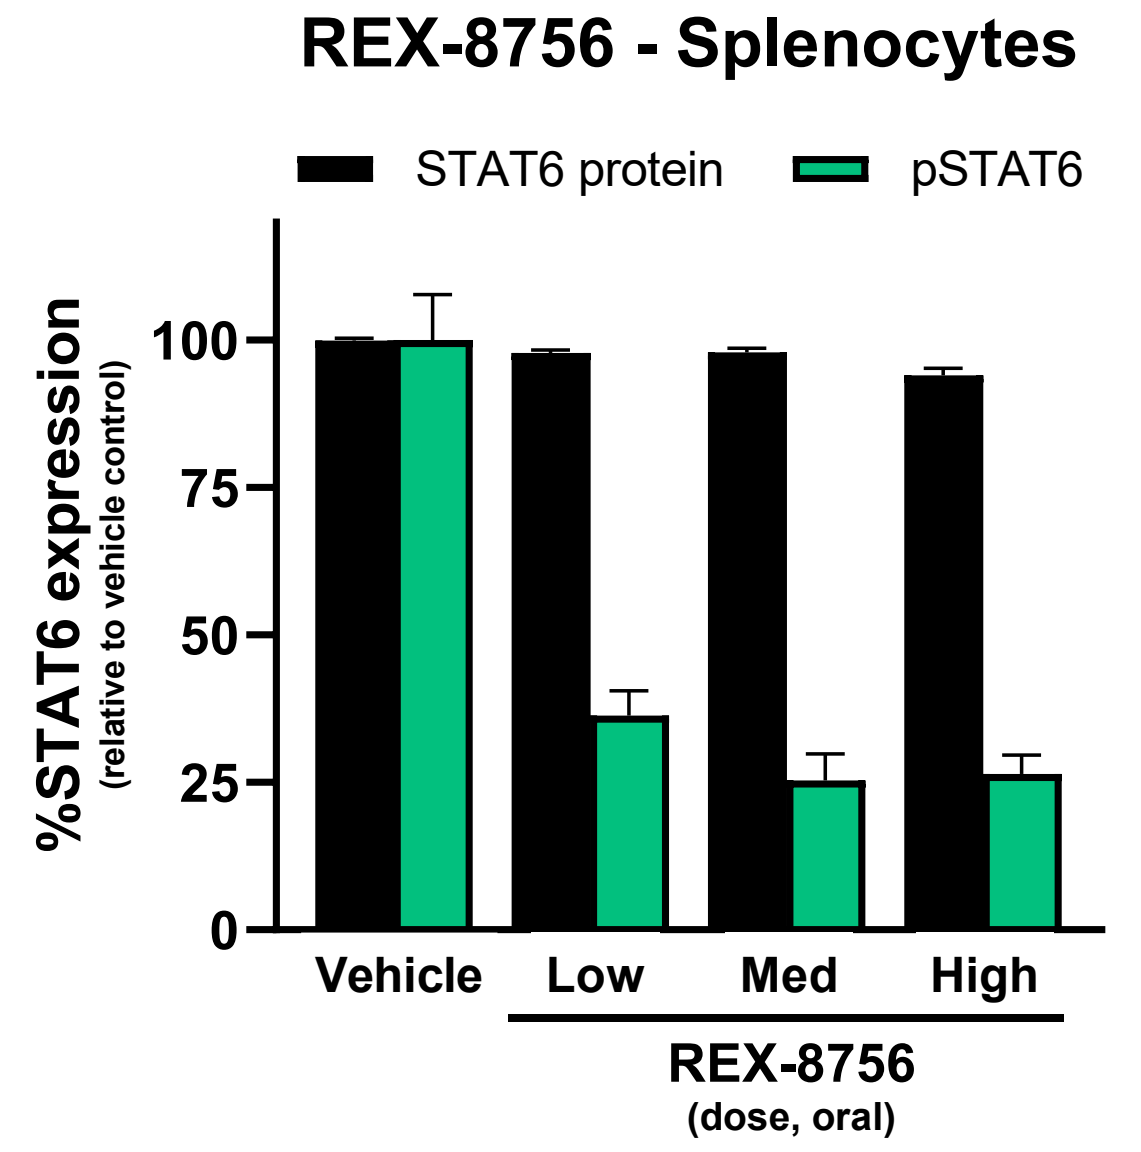
REX-8756 - Splenocytes
STAT6 protein
pSTAT6
%STAT6 expression (relative to vehicle control)
Vehicle
Low
Med
High
REX-8756 (dose, oral)

In a rodent PK/PD study, whole splenocytes were collected 4 hours post a single dose of REX-8756 and stimulated with IL-4 to induce STAT6 activation pathway. Total STAT6 and phospho-STAT6 were quantified by flow cytometry and demonstrated deep and durable pSTAT6 Inhibition without STAT6 protein degradation (Figure 5).

Figure 5. Orally dosed REX-8756 achieves in vivo pSTAT6 inhibition without protein degradation.

CONCLUSIONS

- Orthosteric SH2 domain inhibition may represent an optimal approach to inhibit STAT6
- REX-8756 is a potent and highly selective oral inhibitor of STAT6
- Selectivity of REX-8756 avoids STAT pathways that drive JAK inhibitor safety issues
- REX-8756 demonstrates biologic-like efficacy in a preclinical model of dermatitis
- REX-8756 is currently in a Phase 1a healthy volunteer clinical trial with data expected in 2H 2026

ACKNOWLEDGEMENTS

We would like to thank additional members of the Recludix STAT6 Program Team, including Aryan Alavi, Ksenya Cohen-Katsenelson, Jen Downing, Mark Eckert, Jeremy Hunt, Agnes Kawashima, Brent Marcovitch, Max Orr, Jaime Rodriguez, Allen Sickmier, and John Yeoman for their scientific contributions and feedback on this work.
The STAT6 program is a strategic partnership with Sanofi.

Society for Investigative Dermatology May 13 – 16, 2026, POSTER NUMBER 1055